TRENDS IN PREVALENCE, TREATMENT PATTERNS, MYELOSUPPRESSION, AND BURDEN ON THE HEALTH CARE SYSTEM AMONG PATIENTS WITH SMALL CELL LUNG CANCER: A SEER-MEDICARE ANALYSIS

ROBERT EPSTEIN[1]; JERROD NELMS[2]; DONALD MORAN[3]; CYNTHIA GIRMAN[4]; JOANN KRENITSKY[1]; AND MARC CHIODA[3]

[1] EPSTEIN HEALTH, LLC, WOODCLIFF LAKE, NJ, USA; [2] LUCYNA HEALTH AND SAFETY SOLUTIONS, LLC, LAKELAND, FL, USA; [3] G1 THERAPEUTICS, INC., RESEARCH TRIANGLE PARK, NC, USA; [4] CEROBS CONSULTING, LLC, CHAPEL HILL, NC, USA

INTRODUCTION

- Small cell lung cancer (SCLC), which accounts for approximately 13–17% of all lung cancer cases, is characterized by rapid progression and early, widespread metastases[1,2]
- Standard treatment options for both limited- and extensive-stage SCLC include chemotherapy and radiation therapy[3]
 - Platinum-based combination chemotherapy remains the backbone of first-line therapy, but is now often used in combination with immune checkpoint inhibitors[3]
- Chemotherapy-induced myelosuppression is a frequent complication that may manifest as neutropenia, anemia, and/or thrombocytopenia[4,5]
 - Radiation therapy can also lead to clinically significant myelosuppression, especially when administered in combination with chemotherapy[6,7]
- Consequences of myelosuppression include increased susceptibility to infections, fatigue, bleeding, and sepsis, which can negatively affect patients' quality of life and burden the health care system, owing to the need for supportive care and/or hospitalizations[4,5,8]
- To our knowledge, no real-world study has been conducted in the Surveillance, Epidemiology, and End Results (SEER)-Medicare population to assess the burden of multilineage myelosuppression in patients with SCLC
- Using a SEER-Medicare–linked data set, the current study aimed to estimate the real-world prevalence, map the treatment journey, and quantify the health care resource utilization (HCRU) associated with myelosuppression in patients with limited- and extensive-stage SCLC

OBJECTIVES

- To identify year-over-year trends in the prevalence and treatment of SCLC within a cross section of the US Medicare population
- To quantify and describe the burden of treatment-induced myelosuppression among Medicare patients diagnosed with SCLC

METHODS

DATA SOURCE AND STUDY POPULATION

- This was a retrospective, descriptive study using linked data from 12 SEER registries and the Medicare database
 - SEER-Medicare combines cancer incidence and survival data from US population-based registries (SEER), with insurance claims data from the Medicare program[9,10]
- Patients were aged ≥ 65 years with a new primary diagnosis of SCLC identified from SEER-Medicare data curated between January 2005 and December 2015
- Cancer characteristics, patient survival, and patient demographics were determined using the SEER Patient Entitlement and Diagnosis Summary File
- Treatment patterns and HCRU were derived from Medicare claims data
- Data on the demographics of the general US population aged ≥ 65 years were obtained from the US Census Bureau

SCLC PREVALENCE

- SCLC diagnosis was identified by SEER lung tumor site codes (C34.0–C34.9) and histology codes 8041/3, 8042/3, 8043/3, 8044/3, and 8045/3
- SCLC prevalence (2005–2015) was computed as the number of beneficiaries aged ≥ 65 years with SCLC, divided by the total population aged ≥ 65 years (US Census Bureau)
- Patient demographics (race/ethnicity, sex, and age group) were collected and used to stratify SCLC prevalence

TREATMENT PATTERNS

- Descriptive analysis of the use of chemotherapy, radiation, and surgery across inpatient and outpatient health care settings was performed
 - Treatment types were classified stepwise as first, second, or third line, depending on the temporal sequence in which regimens were prescribed
 - The time span of focus was 2012–2015
- The proportion of patients able to complete their recommended 4–6 cycles of chemotherapy was calculated with an algorithm that factored length of treatment and measurable gaps in treatment

BURDEN OF MYELOSUPPRESSION

- Among patients who received chemotherapy within a given year, the occurrence of myelosuppressive side effects (anemia, thrombocytopenia, neutropenia, pancytopenia) in any medical setting was calculated across the years 2012–2015
- The number and percentage of patients who had ≥ 1 inpatient claim associated with myelosuppression were calculated by study year
- Use of lineage-specific rescue medication (ie, granulocyte colony–stimulating factor [G-CSF], red blood cell [RBC] or platelet transfusions, and erythropoiesis-stimulating agents [ESAs]) among chemotherapy-treated individuals, either before (prophylactic) or after (therapeutic) a myelosuppression event, was tabulated by year

RESULTS

SCLC PREVALENCE

- Across the years 2005–2015, SCLC prevalence peaked in 2009, reaching a maximum of 70.5 per 100,000 persons, and declined thereafter to 48.5 per 100,000 persons in 2015 (**Figure 1**)
 - SCLC-specific mortality rates followed a similar pattern, peaking in 2008, but declining more slowly thereafter
- The cross-sectional SCLC population included slightly more female patients (range: 50.2–52.2%), and patients in this population were predominantly White (range: 86.2–88.9%), with a mean age of 74.2–74.5 years across the study period
- SCLC prevalence was highest among White patients, male patients, and those aged 70–79 years, but year-over-year prevalence of SCLC followed a similar trend across all demographic stratifications

FIGURE 1. MID-YEAR ESTIMATES OF OVERALL PREVALENCE (PER 100,000) OF SCLC AND RELATED MORTALITY: 2005–2015

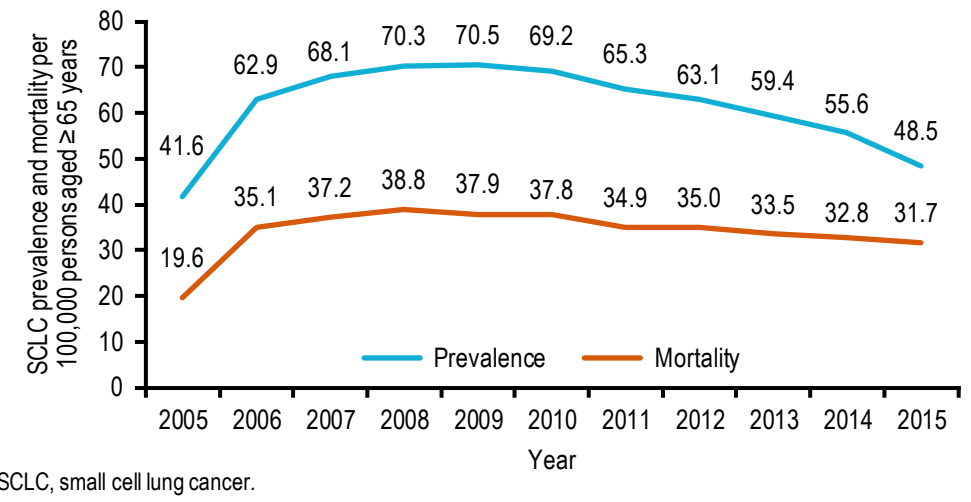
SCLC prevalence and mortality per 100,000 persons aged ≥ 65 years
0
10
20
30
40
50
60
70
80
41.6
62.9
68.1
70.3
70.5
69.2
65.3
63.1
59.4
55.6
48.5
19.6
35.1
37.2
38.8
37.9
37.8
34.9
35.0
33.5
32.8
31.7
Prevalence
Mortality
2005
2006
2007
2008
2009
2010
2011
2012
2013
2014
2015
Year

SCLC, small cell lung cancer.

TREATMENT PATTERNS

- Chemotherapy was the most frequently administered first-line treatment (65.6% of patients), followed by radiation therapy (31.1%); surgery was used less frequently (3.4%; **Figure 2**)
 - 47.9% of patients did not receive second-line treatment, and 98.2% of patients did not receive third-line treatment
- Among patients who received first-line chemotherapy and for whom a second treatment was recorded, most received radiation therapy as second-line or subsequent treatment
 - Likewise, among patients who received first-line radiation therapy and for whom a second treatment was recorded, most received subsequent chemotherapy
- Overall, 57.8% of patients who received chemotherapy completed 4–6 cycles (**Table 1**); this proportion was consistent across the study period (2012–2015; range: 56.9–58.4%)
 - Approximately one-quarter of patients completed only 1 cycle of chemotherapy

FIGURE 2. TREATMENT PATTERNS ACROSS LINES OF THERAPY: 2012–2015

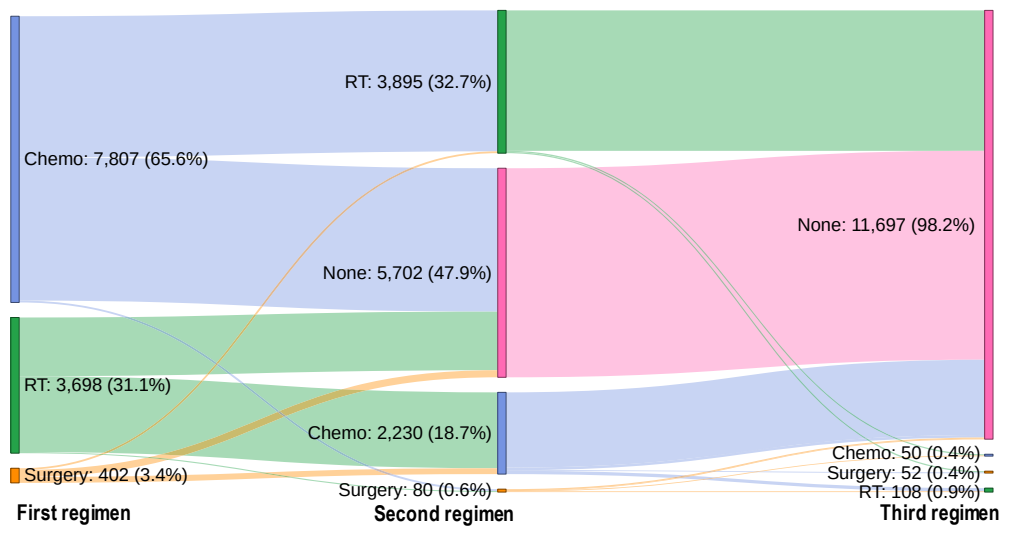
Chemo: 7,807 (65.6%)
RT: 3,698 (31.1%)
Surgery: 402 (3.4%)
RT: 3,895 (32.7%)
None: 5,702 (47.9%)
Chemo: 2,230 (18.7%)
Surgery: 80 (0.6%)
None: 11,697 (98.2%)
Chemo: 50 (0.4%)
Surgery: 52 (0.4%)
RT: 108 (0.9%)
First regimen
Second regimen
Third regimen

For each line of treatment, percentages are calculated using the total number of patients as the denominator.
Chemo, chemotherapy; RT, radiation therapy.

TABLE 1. NUMBER OF CHEMOTHERAPY CYCLES COMPLETED: 2012–2015

Treatment Cycles,[a] n (%)	2012	2013	2014	2015	Overall; 2012–2015
1 cycle	836 (23.4)	531 (23.9)	524 (24.6)	496 (23.6)	2,387 (23.8)
2 cycles	373 (10.4)	194 (8.7)	190 (8.9)	179 (8.5)	936 (9.3)
3 cycles	329 (9.2)	205 (9.2)	177 (8.3)	200 (9.5)	911 (9.1)
4–6 cycles[b]	**2,034 (56.9)**	**1,294 (58.2)**	**1,235 (58.1)**	**1,228 (58.4)**	**5,791 (57.8)**
4 cycles	414 (11.6)	301 (13.5)	283 (13.3)	306 (14.6)	1,304 (13.0)
5 cycles	273 (7.6)	191 (8.6)	192 (9.0)	180 (8.6)	836 (8.3)
6 cycles	1,347 (37.7)	802 (36.1)	760 (35.7)	742 (35.3)	3,651 (36.4)

[a] Chemotherapy regimens were only counted for the year in which the regimen was started.
[b] Guideline-recommended number of chemotherapy cycles.

BURDEN OF MYELOSUPPRESSION

- Almost three-quarters of chemotherapy-treated patients experienced anemia during 2012–2015, varying from 76.8% in 2012 to 62.1% in 2015 (**Table 2**)
- Neutropenia was reported in 45.2% of chemotherapy-treated patients overall; the percentage was generally consistent across 2012–2015
- Thrombocytopenia was reported in 27.0% of chemotherapy-treated patients overall; the percentage was generally consistent across 2012–2015
- Pancytopenia was reported in 24.4% of patients overall, increasing slightly from 22.5% in 2012 to 25.7% in 2015

TABLE 2. OCCURRENCE OF MYELOSUPPRESSION EVENTS: 2012–2015

	2012 (n = 3,588)	2013 (n = 3,509)	2014 (n = 3,427)	2015 (n = 3,434)	Overall; 2012–2015 (N = 13,958)
Occurrence of medical events, %					
Anemia	76.8	76.1	71.3	62.1	71.7
Neutropenia	44.4	46.1	44.1	46.1	45.2
Thrombocytopenia	25.1	27.9	26.6	28.5	27.0
Pancytopenia	22.5	24.2	25.3	25.7	24.4

- During the same period, 74.3% of chemotherapy-treated patients experienced ≥ 1 inpatient admission associated with myelosuppression (**Table 3**)
 - This percentage was generally consistent across 2012–2015
- Hospital admissions for myelosuppression were most frequently reported for anemia (52.8% of patients), followed by neutropenia (33.3%), thrombocytopenia (17.0%), and pancytopenia (14.2%)
- The percentages of patients having ≥ 1 inpatient claim for each type of myelosuppression event were generally consistent from 2012 through 2015, except for anemia, which was less frequent in 2015

TABLE 3. PATIENTS WITH ≥ 1 INPATIENT CLAIM FOR EACH TYPE OF MYELOSUPPRESSION: 2012–2015

	2012 (n = 3,588)	2013 (n = 3,509)	2014 (n = 3,427)	2015 (n = 3,434)	Overall; 2012–2015 (N = 13,958)
Patients with ≥ 1 inpatient claim for myelosuppression, %	77.5	77.6	73.5	68.3	74.3
Anemia	55.9	56.9	52.5	45.7	52.8
Neutropenia	33.6	33.5	31.9	34.0	33.3
Thrombocytopenia	16.3	16.1	16.9	18.8	17.0
Pancytopenia	12.2	13.7	14.9	16.2	14.2

- Among chemotherapy-treated patients, 48.4% of patients received G-CSF, 26.3% received RBC or platelet transfusions, and 11.0% received ESAs (**Table 4**)

TABLE 4. SUPPORTIVE CARE INTERVENTIONS AMONG CHEMOTHERAPY-TREATED PATIENTS WITH SCLC: 2012–2015

Resource/Treatment, %	2012 (n = 3,588)	2013 (n = 3,509)	2014 (n = 3,427)	2015 (n = 3,434)	Overall; 2012–2015 (N = 13,958)
G-CSF	52.3	52.4	44.1	44.6	48.4
RBC or platelet transfusion	27.7	26.9	26.3	24.4	26.3
ESA usage	13.4	12.1	10.3	8.0	11.0

ESA, erythropoiesis-stimulating agent; G-CSF, granulocyte colony–stimulating factor; RBC, red blood cell; SCLC, small cell lung cancer.

CONCLUSIONS

- Across 2005–2015, SCLC prevalence peaked in 2009 and then declined, following a similar temporal pattern across all races/ethnicities, sexes, and age groups
- Chemotherapy remains the cornerstone of treatment for SCLC, alongside radiation therapy
- Only 57.8% of the SEER-Medicare patients with SCLC who were studied completed guideline-recommended 4–6 cycles of chemotherapy
 - This shortfall underscores the frailty of patients experiencing this aggressive disease
- Myelosuppression imposes a substantial burden on older patients with SCLC

Virtual ISPOR Annual Meeting | May 17–20, 2021

ACKNOWLEDGMENTS:
Study sponsored by G1 Therapeutics, Inc. Medical writing assistance was provided by Alligent Europe (Envision Pharma Group), funded by G1 Therapeutics, Inc. We thank Huan Huang (G1 Therapeutics, Inc.) for her assistance with poster development.

REFERENCES:

1. Howlader N, et al. SEER Cancer Statistics Review (CSR) 1975-2017. https://seer.cancer.gov/csr/1975_2017/. Accessed March 2021.
2. Wang S, et al. *Sci Rep*. 2017;7:1339.
3. PDQ Adult Treatment Editorial Board. Small Cell Lung Cancer Treatment (PDQ®)—Health Professional Version. https://www.cancer.gov/types/lung/hp/small-cell-lung-treatment-pdq. Accessed April 2021.
4. Carey PJ. *Drug Saf*. 2003;26:691–706.
5. Barreto JN, et al. *J Pharm Pract*. 2014;27:440–6.
6. Barney CL, et al. *Int J Radiat Oncol Biol Phys*. 2018;100:748–55.
7. Faivre-Finn C, et al. *Lancet Oncol*. 2017;18:1116–25.
8. Epstein RS, et al. *Adv Ther*. 2020;37:3606–18.
9. Warren JL, et al. *Med Care*. 2002;40:IV-3-18.
10. Enewold L, et al. *J Natl Cancer Inst Monogr*. 2020;2020:3–13.